Preclinical pharmacokinetic (PK) and tumour growth inhibition (TGI) modelling for mANK-101, an anchored murine interleukin-12 (IL-12) complex for intratumoural administration for solid cancer

Hitesh Mistry[1], David Hodson[1*], David Orrell[1], Sailaja Battula[2], Leisha A. Emens[2], Howard L. Kaufman[2], Michael M Schmidt[2], Christophe Chassagnole[1]

[1]Physiomics plc, The Oxford Science Park, Oxford, United Kingdom. [2]Ankyra Therapeutics, Cambridge, MA, USA

*Tel. +44 (0)1865 784980. Email: dhodson@physiomics.co.uk

Abstract No A141, AACR-NCI-EORTC – October 11-14 2023

Introduction

IL-12 is a pro-inflammatory cytokine capable of inducing a robust anti-tumour immune response. However, issues with systemic toxicity have limited its potential for use within a clinical setting.

Ankyra Therapeutics has developed a novel anchored drug delivery platform that locally retains immune agonists such as IL-12 following intratumoural (IT) delivery. ANK-101 and its murine surrogate mANK-101 consist of Alhydrogel® scaffolds anchored to genetic fusions of IL-12 and Alum-binding peptide (ABP).

This work describes a model-based assessment of the mechanism of action in CT26 models, as well as the development of a PK-Tumour Growth Inhibition (TGI) model that describes all studies in the CT26 syngeneic mouse model.

Material and methods

Model-based assessment of TGI. Tumour growth inhibition was assessed in CT26 syngeneic tumour models. In order to assess if a treatment effect exists, the model is first fit to the data assuming there is no treatment effect,

$$M1\colon R_{ij} = R_0 + g_i * t_{ij} + e_{ij}$$

R_{ij} is the radius at time i for mouse j and is equal to the initial R_0, assumed to be the same for all mice. g_i, assumed to be normally distributed with unknown mean and variance represents the tumour growth rate for mouse i. Residual error, e_{ij}, is the unexplained variance in the data and is assumed to be normally distributed with mean 0 and unknown variance.

To assess if there is a treatment effect, the likelihood ratio-test (LRT) is used to compare the above model to M2,

$$M2\colon R_{ij} = R_0 + (g_i + d1 * mIL12 + d2 * mANK101) * t_{ij} + e_{ij}$$

Where $d1$ and $d2$ are treatment effect parameters representing the injection of free murine IL-12 and mANK-101 respectively.

Model-based biomarker validation. The relationship between observed biomarker estimates and tumour growth rate after administration of mANK-101 was performed by comparing the %CV (coefficient of variance of the growth rate distribution) with/without any covariates between model M1 and M3.

$$M1\colon R_{ij} = R_0 + g_i * t_{ij} + e_{ij}$$

$$M3\colon R_{ij} = R_0 + (g_i + a_0 * Covariate_i) * t_{ij} + e_{ij}$$

Where $Covariate_i$ is the biomarker value of mouse i. Models were compared for statistical significance using the LRT, and assessing how the covariate impacted the coefficient of variation (CV%) of the baseline tumour growth rate.

PK-TGI Model. The following model was fitted to efficacy study data assessing the role of mANK-101 on CT26 syngeneic tumour models.

$$\frac{dD_{mIL12}}{dt} = -k_{a,1}D_{mIL12}$$

$$\frac{dC}{dt} = k_{a,1}D_{mIL12} + k_{21}P - (k_{12}+k_e)C$$

$$\frac{dP}{dt} = -k_{21}P + k_{12}C$$

$$Plasma\ Concentration(t) = C/(V/F_1)$$

$$\frac{dI_0}{dt} = \frac{-k_{act} \cdot I_0 \cdot C \cdot F_1}{V}$$

$$\frac{dI_A}{dt} = \frac{k_{act} \cdot I_0 \cdot C \cdot F_1}{V}$$

$$\frac{dR}{dt} = g - d\frac{I_A}{EC50 + I_A}R$$

The PK model was derived from Momin *et al.* [1], where D_{mIL12} represents IL-12 injected into the tumour (either using mANK-101 or murine IL-12), C represents the central compartment, and P represents the peripheral compartment. The TGI model assumes that there is a finite immune response which is recruited that is proportional to the concentration of IL-12 within the plasma (I_0) The activated response I_A is capable of inhibiting the growth rate of the tumour (R) at rate d and a given affinity $EC50$. PK parameters were fitted initially, and fixed when fitting TGI parameters k_{act}, g, d and $EC50$.

Model-based assessment of TGI

Model-based assessment of TGI confirms a significant reduction in tumour growth rate of CT26 tumours after administration of mANK-101.

The raw TGI data (faint-lines individual mice and darker lines represent the mean) from the biomarker study is shown in Figure 1 (left-panel) for single dose of mIL12 and mANK-101. The comparison between M1 v M2 gave a LRT p-value of 0.002, indicating we can reject the null hypothesis that there is no treatment effect. Figure 1 (right-panel) indicates that the model fits the data well.

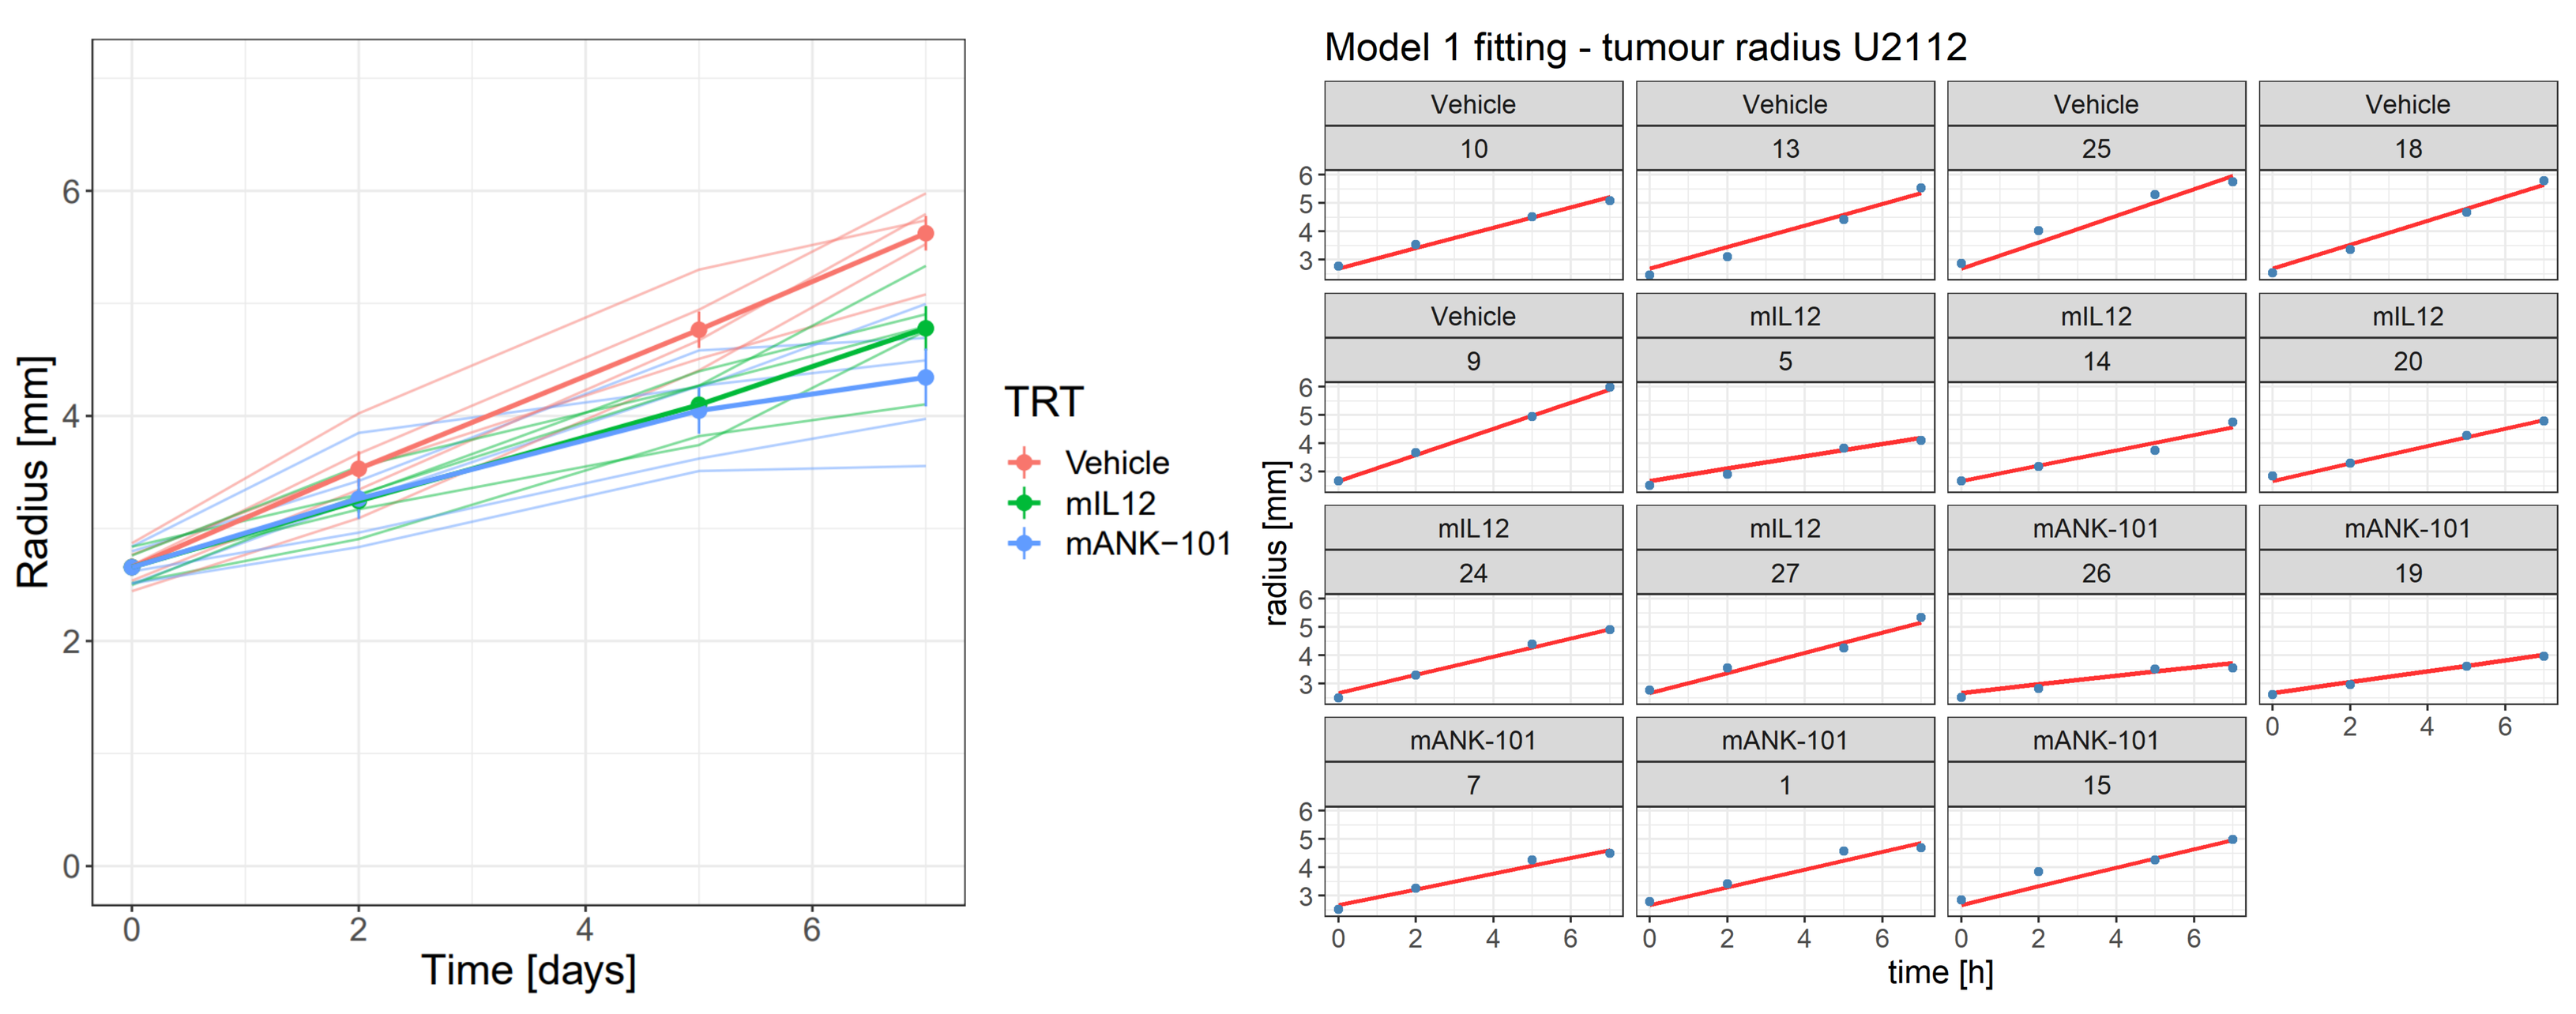
Radius [mm]
Time [days]
TRT
Vehicle
mIL12
mANK−101
Model 1 fitting - tumour radius U2112
radius [mm]
time [h]

Figure 1. *Left-panel*) Plot of the tumour growth data from the CT-26 biomarker study – volumes converted to radius by mapping volumes onto a sphere. *Right-panel*) Plot showing the individual fits (solid red-line) to the raw tumour size data (blue-dots) for the CT26 biomarker study.

Model-based biomarker validation

Highlighted in red boxes (Figure 2) are biomarkers which show visually a marked difference in both treated arms versus control at day 7. Assessment of how the variance in growth rate can be explained by single biomarkers was then performed. The subsequent analyses (Table 1) were limited to assessment of control and mANK-101 treated cohorts. Numerous markers reduced the %CV compared to the base model. Of all the markers, %CD8 cells explained most of the variance, which is consistent with the goal of the treatment - to further potentiate the adaptive immune response within these tumours.

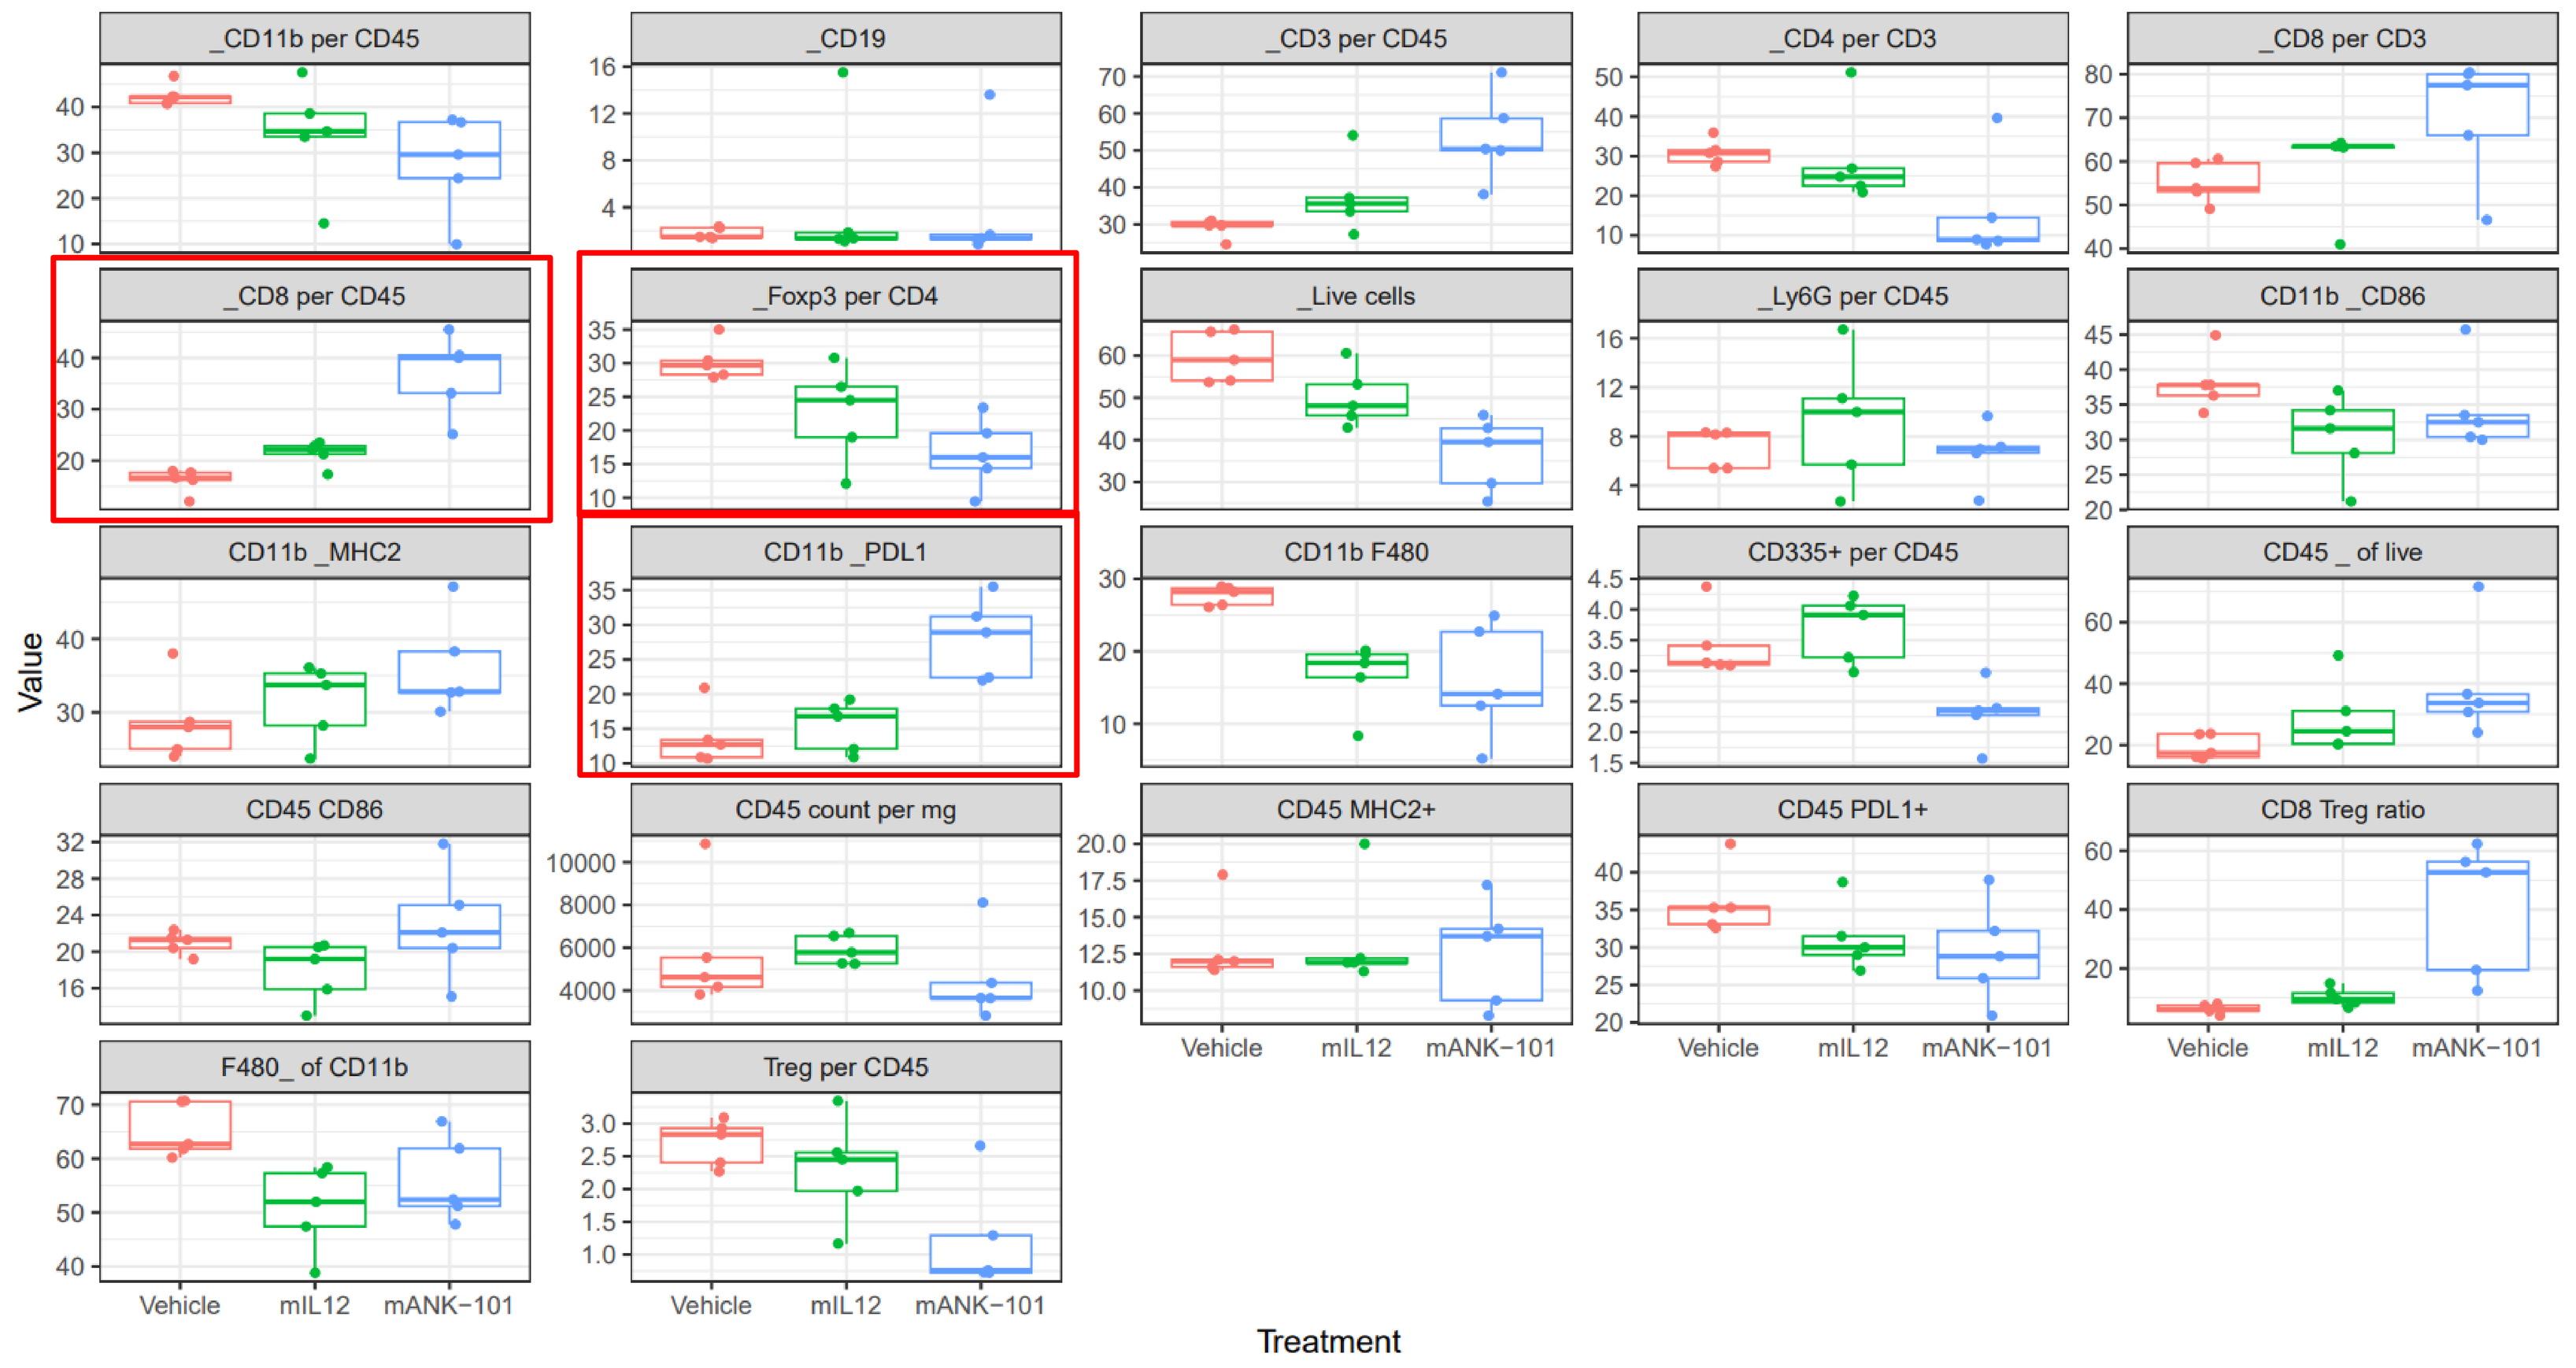
_CD11b per CD45
_CD19
_CD3 per CD45
_CD4 per CD3
_CD8 per CD3
_CD8 per CD45
_Foxp3 per CD4
_Live cells
_Ly6G per CD45
CD11b _CD86
CD11b _MHC2
CD11b _PDL1
CD11b F480
CD335+ per CD45
CD45 _ of live
CD45 CD86
CD45 count per mg
CD45 MHC2+
CD45 PDL1+
CD8 Treg ratio
F480_ of CD11b
Treg per CD45
Value
Vehicle
mIL12
mANK−101
Treatment

Figure 2. Plots comparing tumour-infiltrating biomarkers as measured by flow cytometry for the 3 treatment groups in the CT26 model.

Table 1. Results of assessing the correlation of biomarkers to treatment effect in the CT26 model.

Biomarker	LRT p-value	%CV Base	%CV with Biomarker
%CD8 per CD3	0.027	28.5	13
%CD8 per CD45	0.002	28.5	13.4
CD11b/PDL1	0.004	28.5	14.2
CD45 CD86	0.237	28.5	19
CD45 % of live	0.141	28.5	21.6
%Ly6G per CD45	0.471	28.5	24.9
CD19	0.854	28.5	28.2

PK-TGI model

The PK-TGI model was able to successfully capture the growth delay induced by mANK-101 in CT26 syngeneic tumours. The model (solid black line) was found to describe all the data (coloured dots) equally well (Figure 3). The parameter values were estimated with good precision (Table 2).

Table 2. Final parameter values for the TGI model. CI – confidence interval

Parameters	Units	Estimate (95%CI)
k_{act}	L/μg	0.10 (0.06, 1.63)
g	mm/day	0.47 (0.44, 0.50)
d	1/day	0.12 (0.11, 0.13)
$EC50$	%	5.4 (3.2, 9.1)

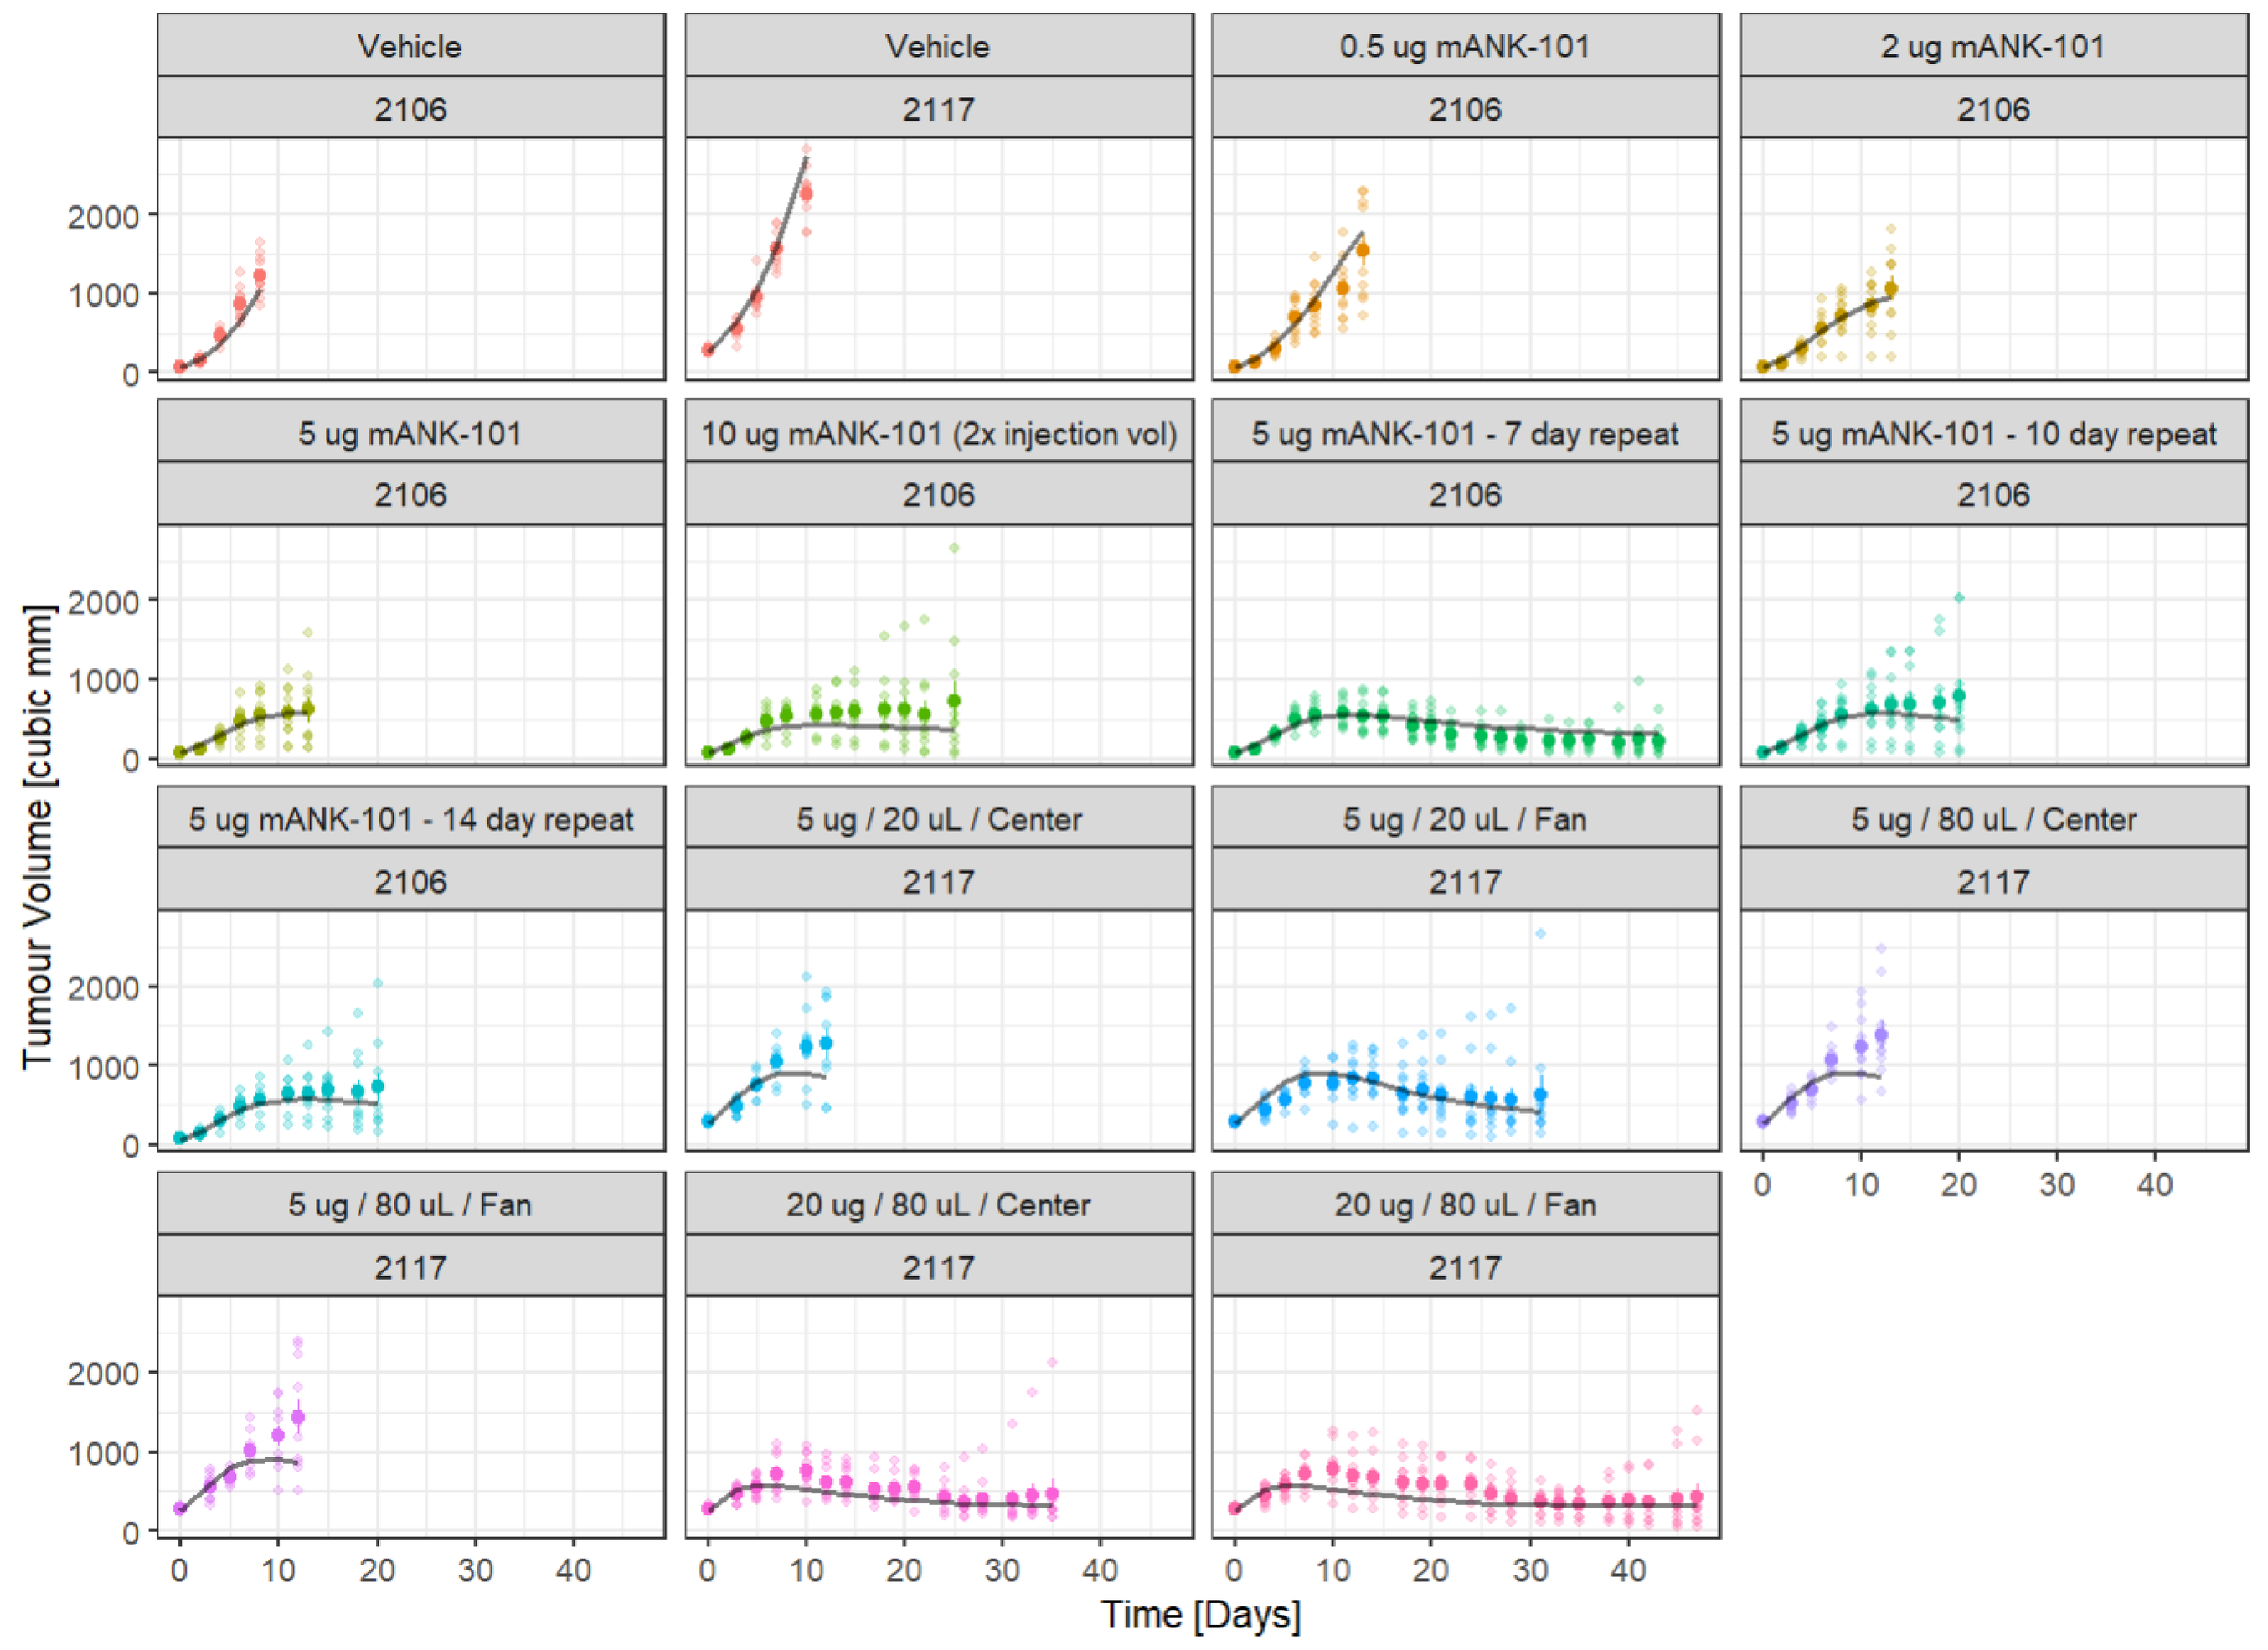
Vehicle 2106
Vehicle 2117
0.5 ug mANK-101 2106
2 ug mANK-101 2106
5 ug mANK-101 2106
10 ug mANK-101 (2x injection vol) 2106
5 ug mANK-101 - 7 day repeat 2106
5 ug mANK-101 - 10 day repeat 2106
5 ug mANK-101 - 14 day repeat 2106
5 ug / 20 uL / Center 2117
5 ug / 20 uL / Fan 2117
5 ug / 80 uL / Center 2117
5 ug / 80 uL / Fan 2117
20 ug / 80 uL / Center 2117
20 ug / 80 uL / Fan 2117
Tumour Volume [cubic mm]
Time [Days]

Figure 3. Plots showing the model fits (black lines) to all the TGI data (coloured dots/lines) across all CT26 data provided.

Conclusions

A PK-TGI model that describes all the PK and TGI data available for mANK-101 across numerous different studies was developed. The final model was able to describe all the efficacy studies done with different doses, schedules, injection sites and starting volumes well. The PK modelling highlighted the lower systemic bioavailability of alum anchored mIL12/ABP versus free mIL12/ABP and also highlighted that for alum anchored mIL12/ABP the absorption rate into plasma is significantly slower than its elimination from plasma. Furthermore, the biomarker analysis conducted is consistent with a mechanism of drug action of tumour cell death mediated by IL-12 mediated T-cell activation.

References:

[1] Momin, N., Palmeri, J.R., Lutz, E.A. et al. Maximizing response to intratumoral immunotherapy in mice by tuning local retention. Nat Commun 13, 109 (2022). https://doi.org/10.1038/s41467-021-27390-6

Physiomics plc ©2023

www.physiomics.co.uk